The Prenylation Inhibitor Lonafarnib Can Induce Post-treatment ALT Flares with Viral Clearance in Patients with Chronic Delta Hepatitis

C. Yurdaydin[1*], R. Idilman[1], C. Kalkan[1], F. Karakaya[1], A. Caliskan[1], E. Karatayli[1], S. Karatayli[1], A. Mithat Bozdayi[1], C. Koh[2], T. Heller[2], J. S. Glenn[3*]

Division of Gastroenterology, University of Ankara Medical School, Ankara, Turkey[1]; Liver Diseases Branch, National Institute of Diabetes & Digestive & Kidney Diseases, National Institutes of Health, Bethesda, Maryland[2]; Division of Gastroenterology and Hepatology, Stanford University School of Medicine[3]. *These authors share senior authorship.

1. Abstract

Background/Aims: Chronic delta hepatitis (CDH) is the most severe form of viral hepatitis. There is currently no approved therapy for CDH. The only established treatment is with interferons, effective in only 25 to 30% of patients. New treatment options are needed in CDH. The prenylation inhibitor lonafarnib (LNF) is the first pharmacological investigational treatment specific for hepatitis D virus (Koh et al, Lancet Infect Dis 2015). Here we report for the first time post-treatment ALT flares and their outcomes in patients treated with 12 or 24 wks of LNF in various treatment regimens.

Methods: 27 patients were analyzed who had detectable HDV RNA after receiving LNF for 12 or 24 wks in the LOWR HDV-1 and LOWR HDV-2 trials.

A post-treatment ALT flare was defined as elevation of ALT to >2x baseline ALT level

Results: To date, 5 of 27 (18.5%) patients have experienced post-treatment ALT flares. These post-treatment flares (median ALT 190 U/mL, range 110-1355 U/mL) led to ALT normalization and HDV RNA became negative within 12-24 wks. These patients came from a variety of LNF treatment cohorts: LNF 200mg bid, 12 wks; LNF 300 mg bid, 12 wks, LNF 100mg bid/RTV 50mg bid, 12 wks; LNF 75 mg bid/RTV 100mg bid, 12 wks, followed by addition of pegylated interferon alfa (PEG IFN α) for 12 wks; LNF 50mg bid/RTV 100mg bid, 24 wks. One patient cleared HBV DNA and subsequently cleared HBsAg; the others did not, although 2 other patients exhibited declines in HBV DNA of 2 logs or greater. All five patients exhibited rapid initial declines of HDV RNA with initiation of LNF that were eventually followed by more gradual rises on therapy associated with decreased LNF exposure (due to dose reductions or excessive GI side effects).

Conclusion: The data suggest that a short course of LNF may contribute to an effective reset and activation of the immune reactivity in CDH, which in some cases my spread to HBV. Thus, there appear to be at least two pathways for achieving HDV negativity with LNF therapy: On treatment LNF-induced progressive suppression to HDV negativity with ALT normalization (more classical antiviral approach, e.g. exemplified in the ongoing LOWR 2 study), and LNF-induced post-treatment anti-HDV ALT flares (described here). The mechanisms of this latter immune restoration are being explored and may lead to the prospective identification of patients who are likely to experience this remarkable outcome.

2. Background - Lonafarnib

- Lonafarnib is a small molecule, oral, prenylation inhibitor
- Well-characterized through Phase 3
 - >2,000 patients dosed in oncology program by Merck
 - Dose limiting toxicity is GI (class effect)
- LOWR HDV (<u>LO</u>nafarnib <u>W</u>ith <u>R</u>itonavir in <u>HDV</u>) Program
 - Identify dose and regimen for registration study
- Over 100 HDV patients dosed across 3 international sites

NIH* "POC" Study N = 14	LOWR HDV – 1 "Combinations" Study N = 15	LOWR HDV – 2 "Dose-Finding" Study N = 46

LOWR HDV – 3 "QD" Study N = 21	LOWR HDV – 4 "Dose-Escalation" Study N = 15

3. Observations

- Post-treatment ALT flares observed from multiple LNF treatment cohorts
- After ≥ 12 weeks LNF treatment, in LOWR HDV – 1 and – 2 studies:
 - 27 patients with ≥ 24 weeks post-treatment follow-up
 - 5 of 27 (18.5%) patients have experienced post-treatment ALT flares
 - 5 of 5 (100%) with post-treatment HDV-RNA PCR-negativity
 - 4 of 5 (80%) normalized ALT; 1 normalizing
 - 5 of 5 (100%) suppressed HBV-DNA
- Post-treatment ALT flares followed by viral clearance
 - Novel observation (vs. prior IFN studies) in HDV
- LNF treatment may reset / activate the immune response to HDV and HBV

4. Results: HDV-RNA PCR-negative Following ALT Flare

Patient 1
LNF 200 mg BID

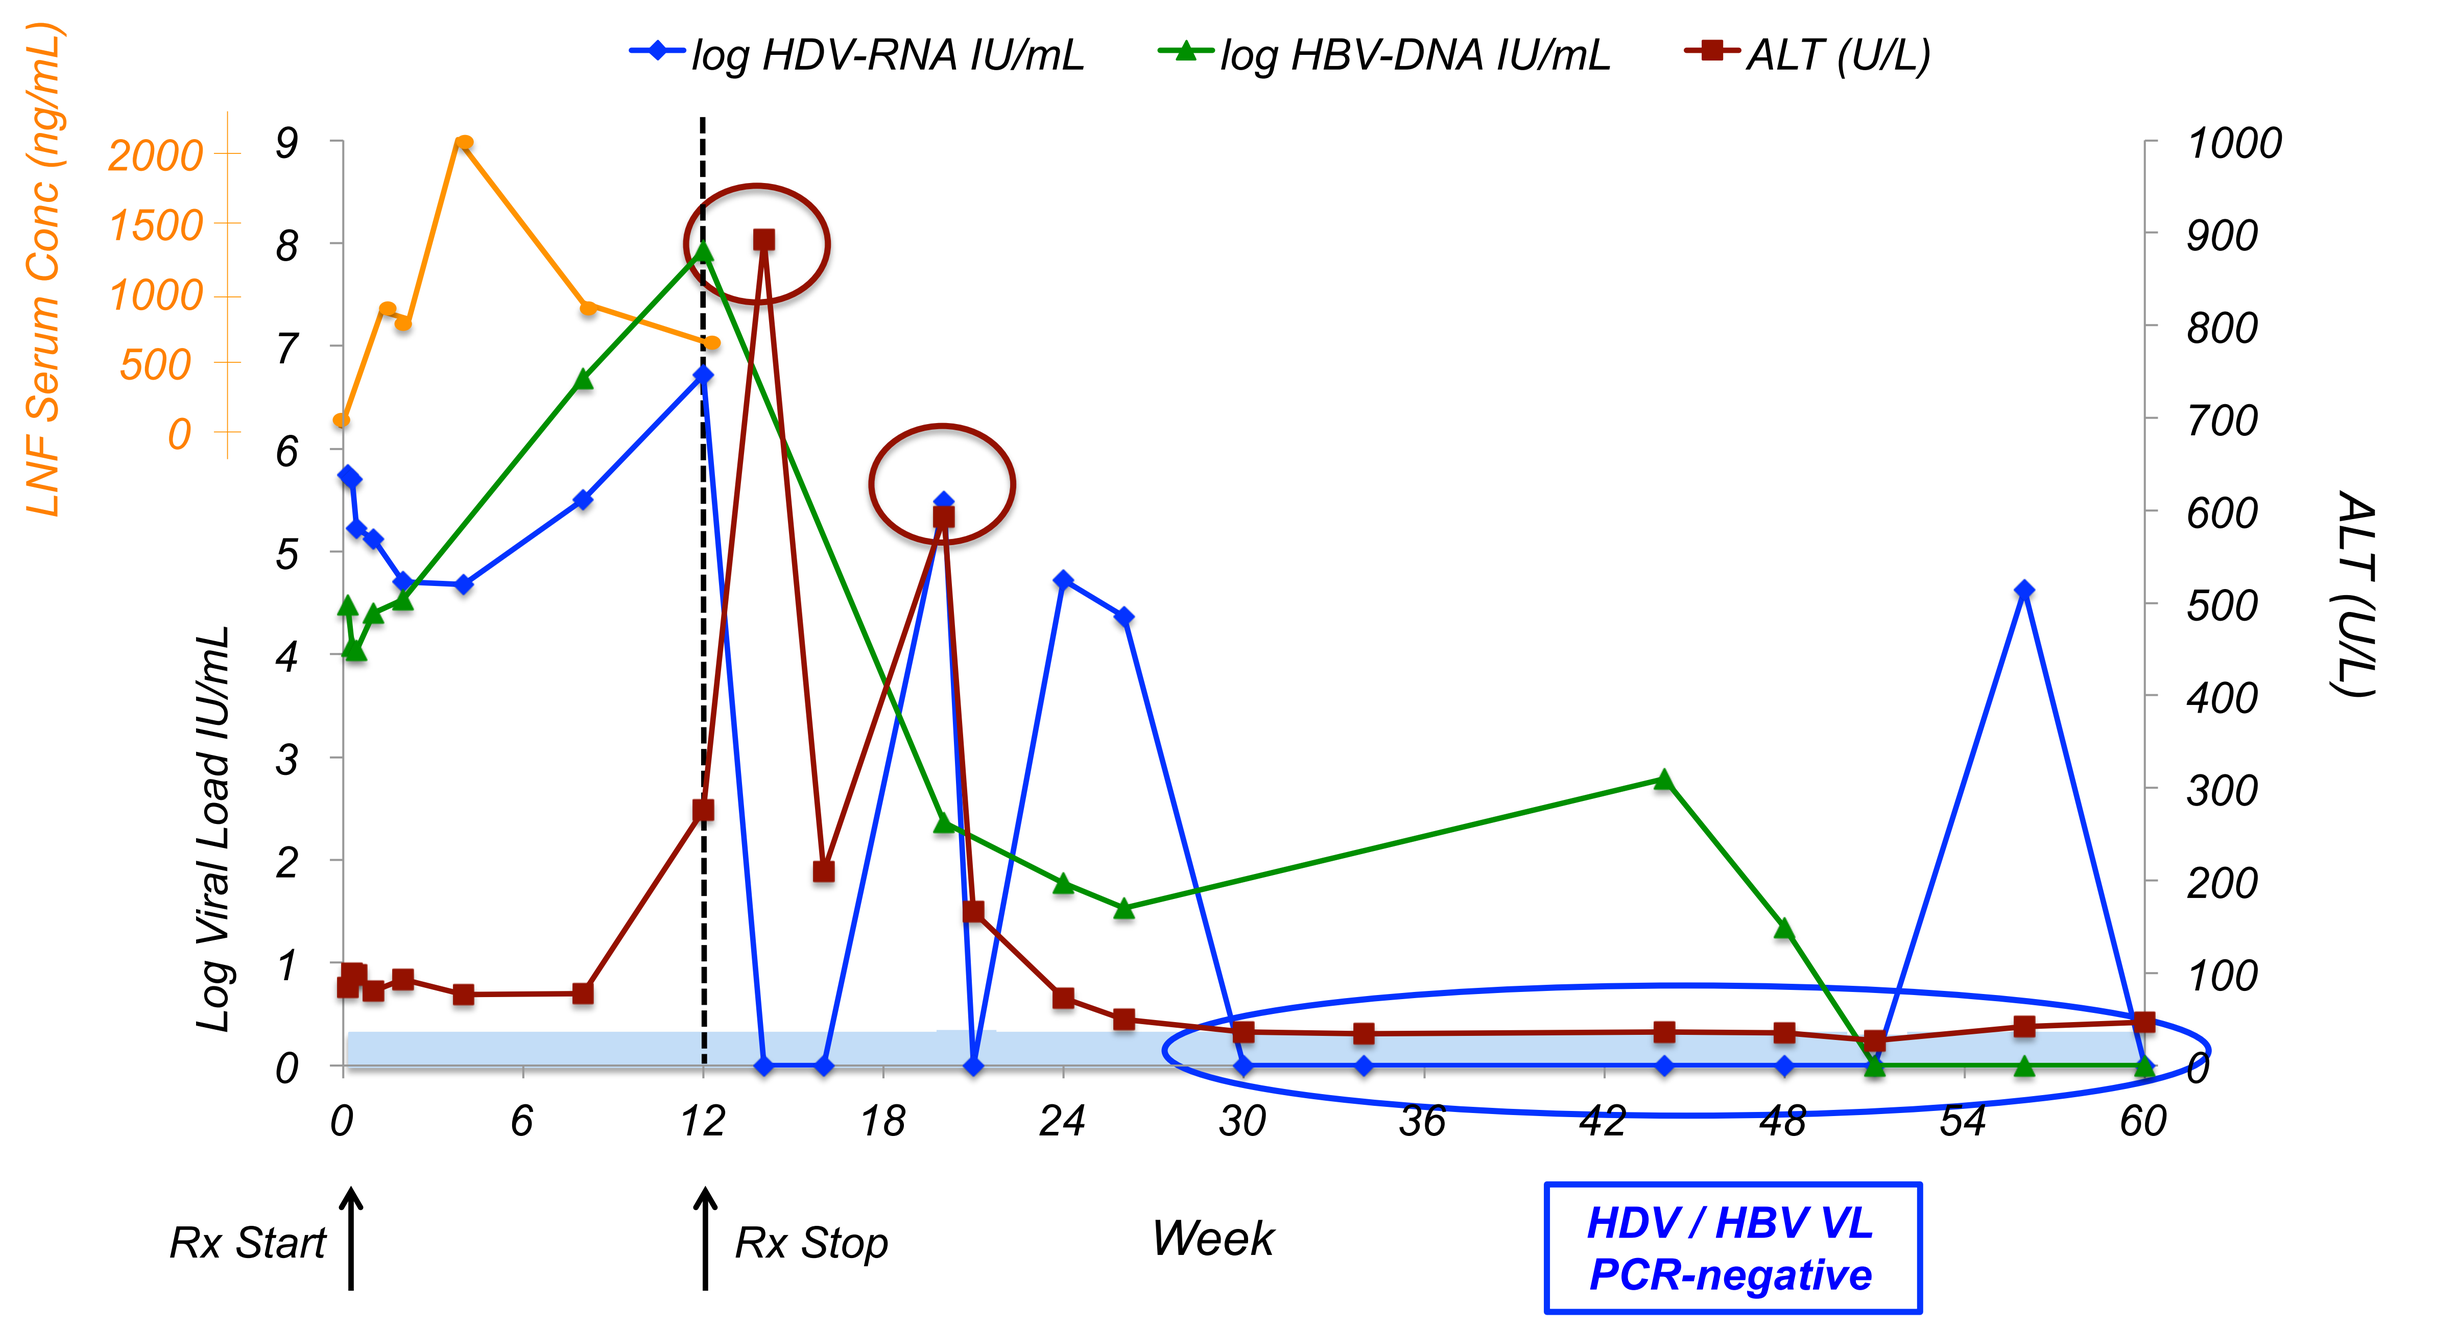

log HDV-RNA IU/mL
log HBV-DNA IU/mL
ALT (U/L)
LNF Serum Conc (ng/mL)
Log Viral Load IU/mL
ALT (U/L)
Week
Rx Start
Rx Stop
HDV / HBV VL PCR-negative

Patient 2
LNF 100 mg BID + RTV 50 mg BID

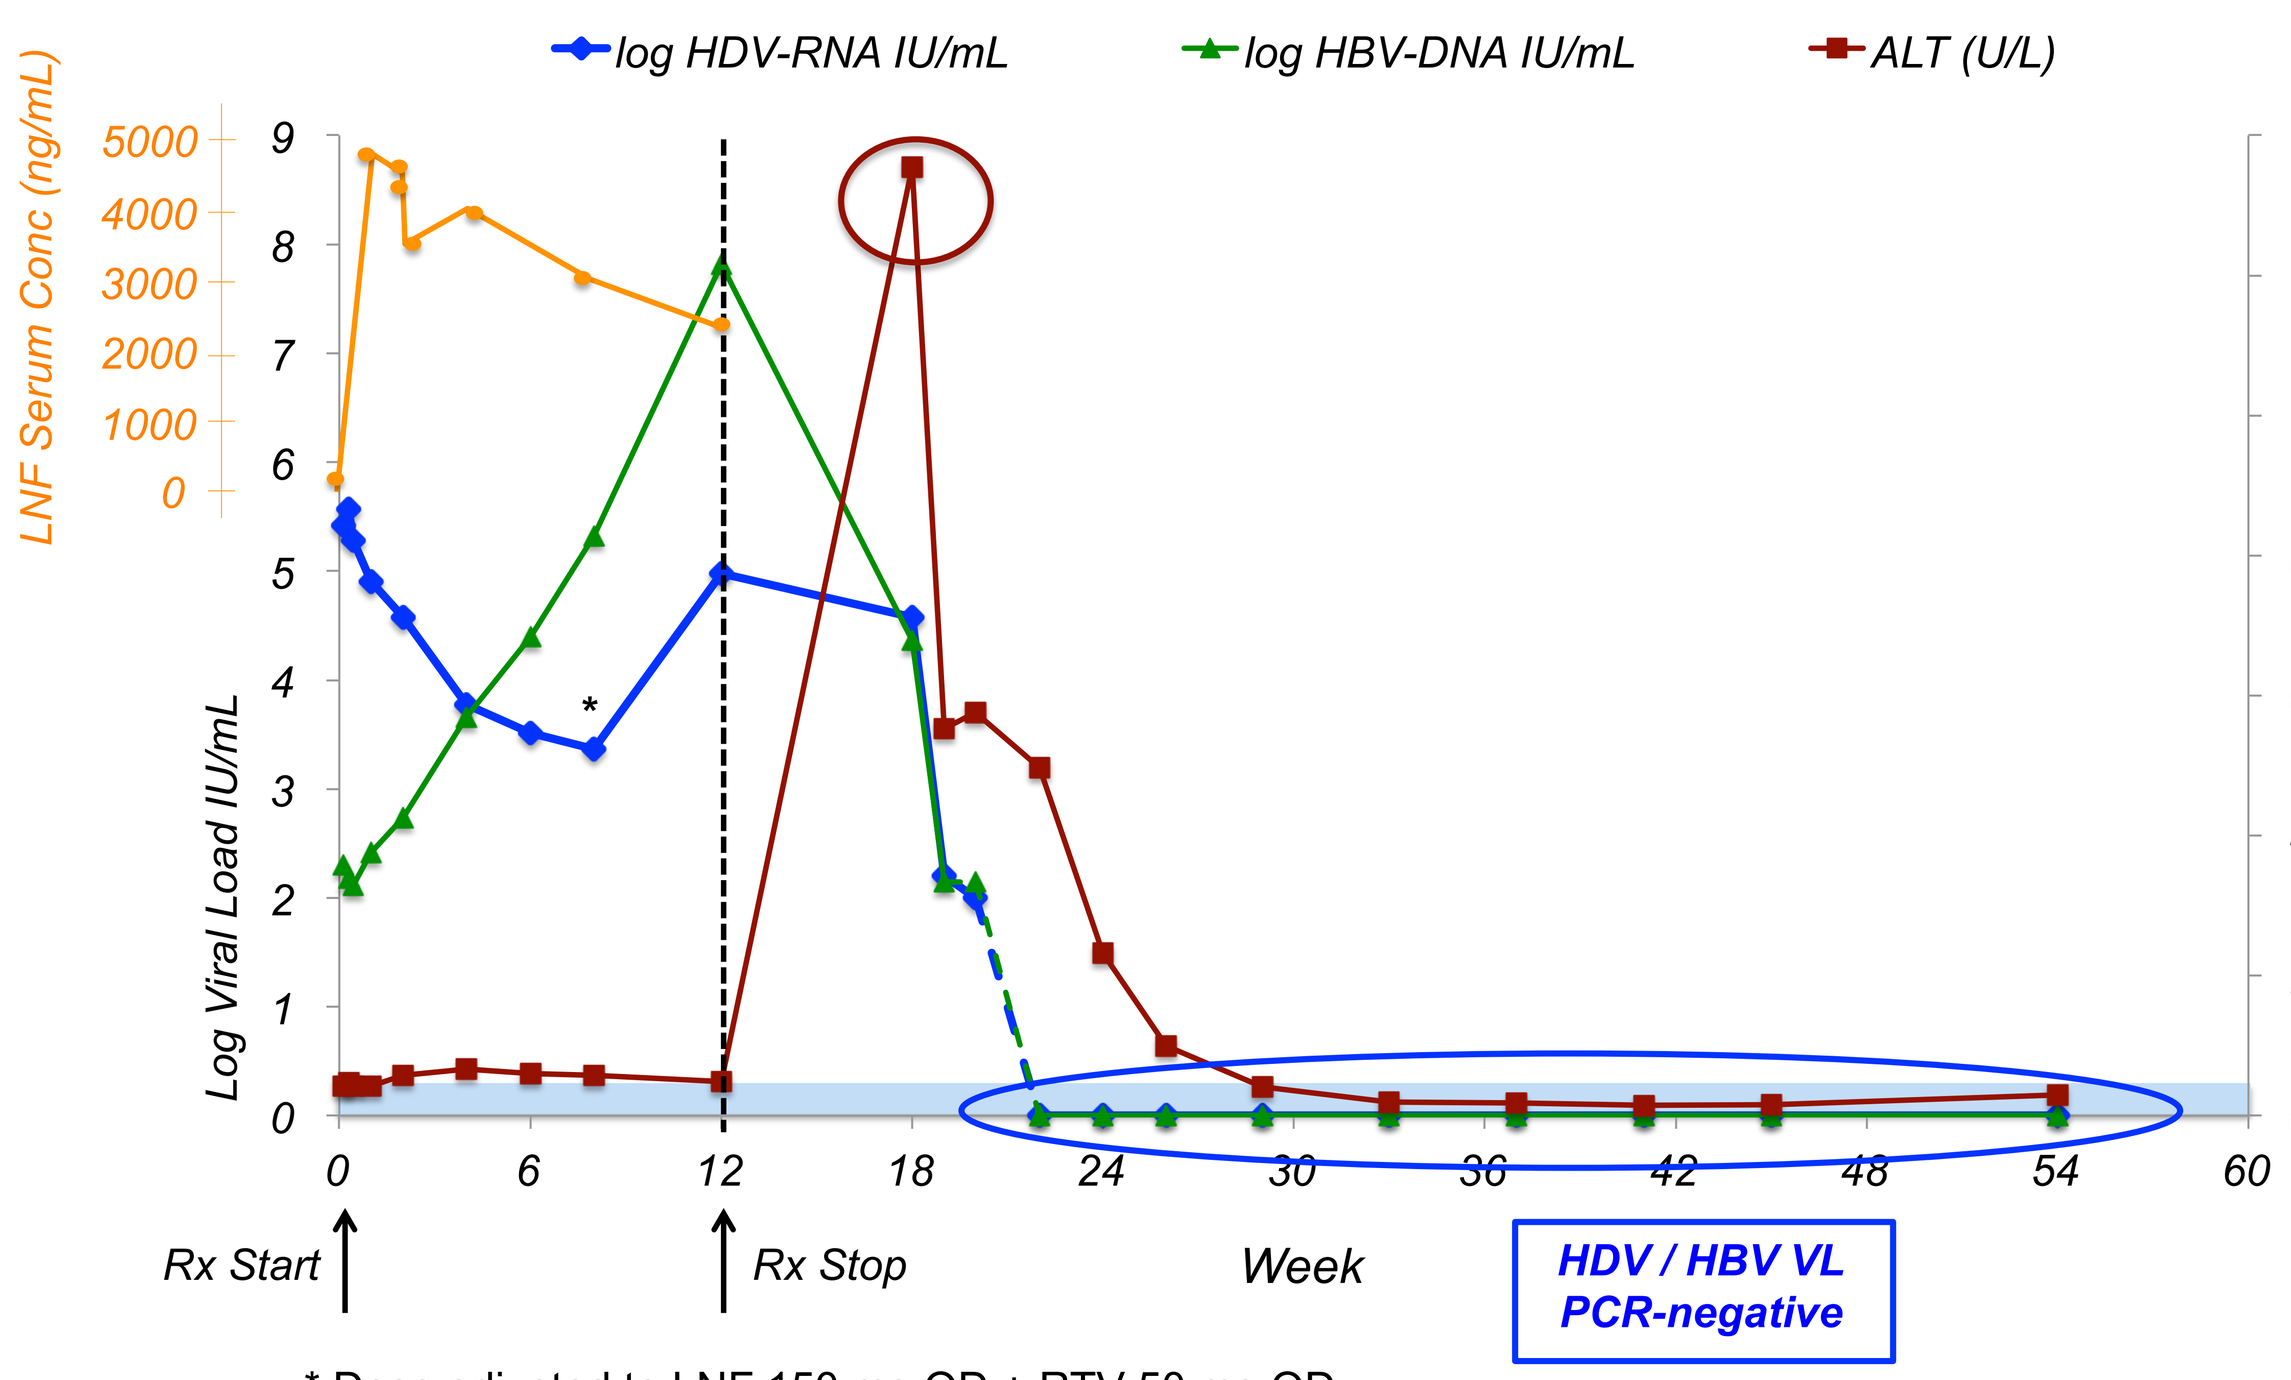

log HDV-RNA IU/mL
log HBV-DNA IU/mL
ALT (U/L)
LNF Serum Conc (ng/mL)
Log Viral Load IU/mL
ALT (U/L)
Week
Rx Start
Rx Stop
HDV / HBV VL PCR-negative

* Dose adjusted to LNF 150 mg QD + RTV 50 mg QD

Patient 3
LNF 50 mg BID + RTV 100 mg BID

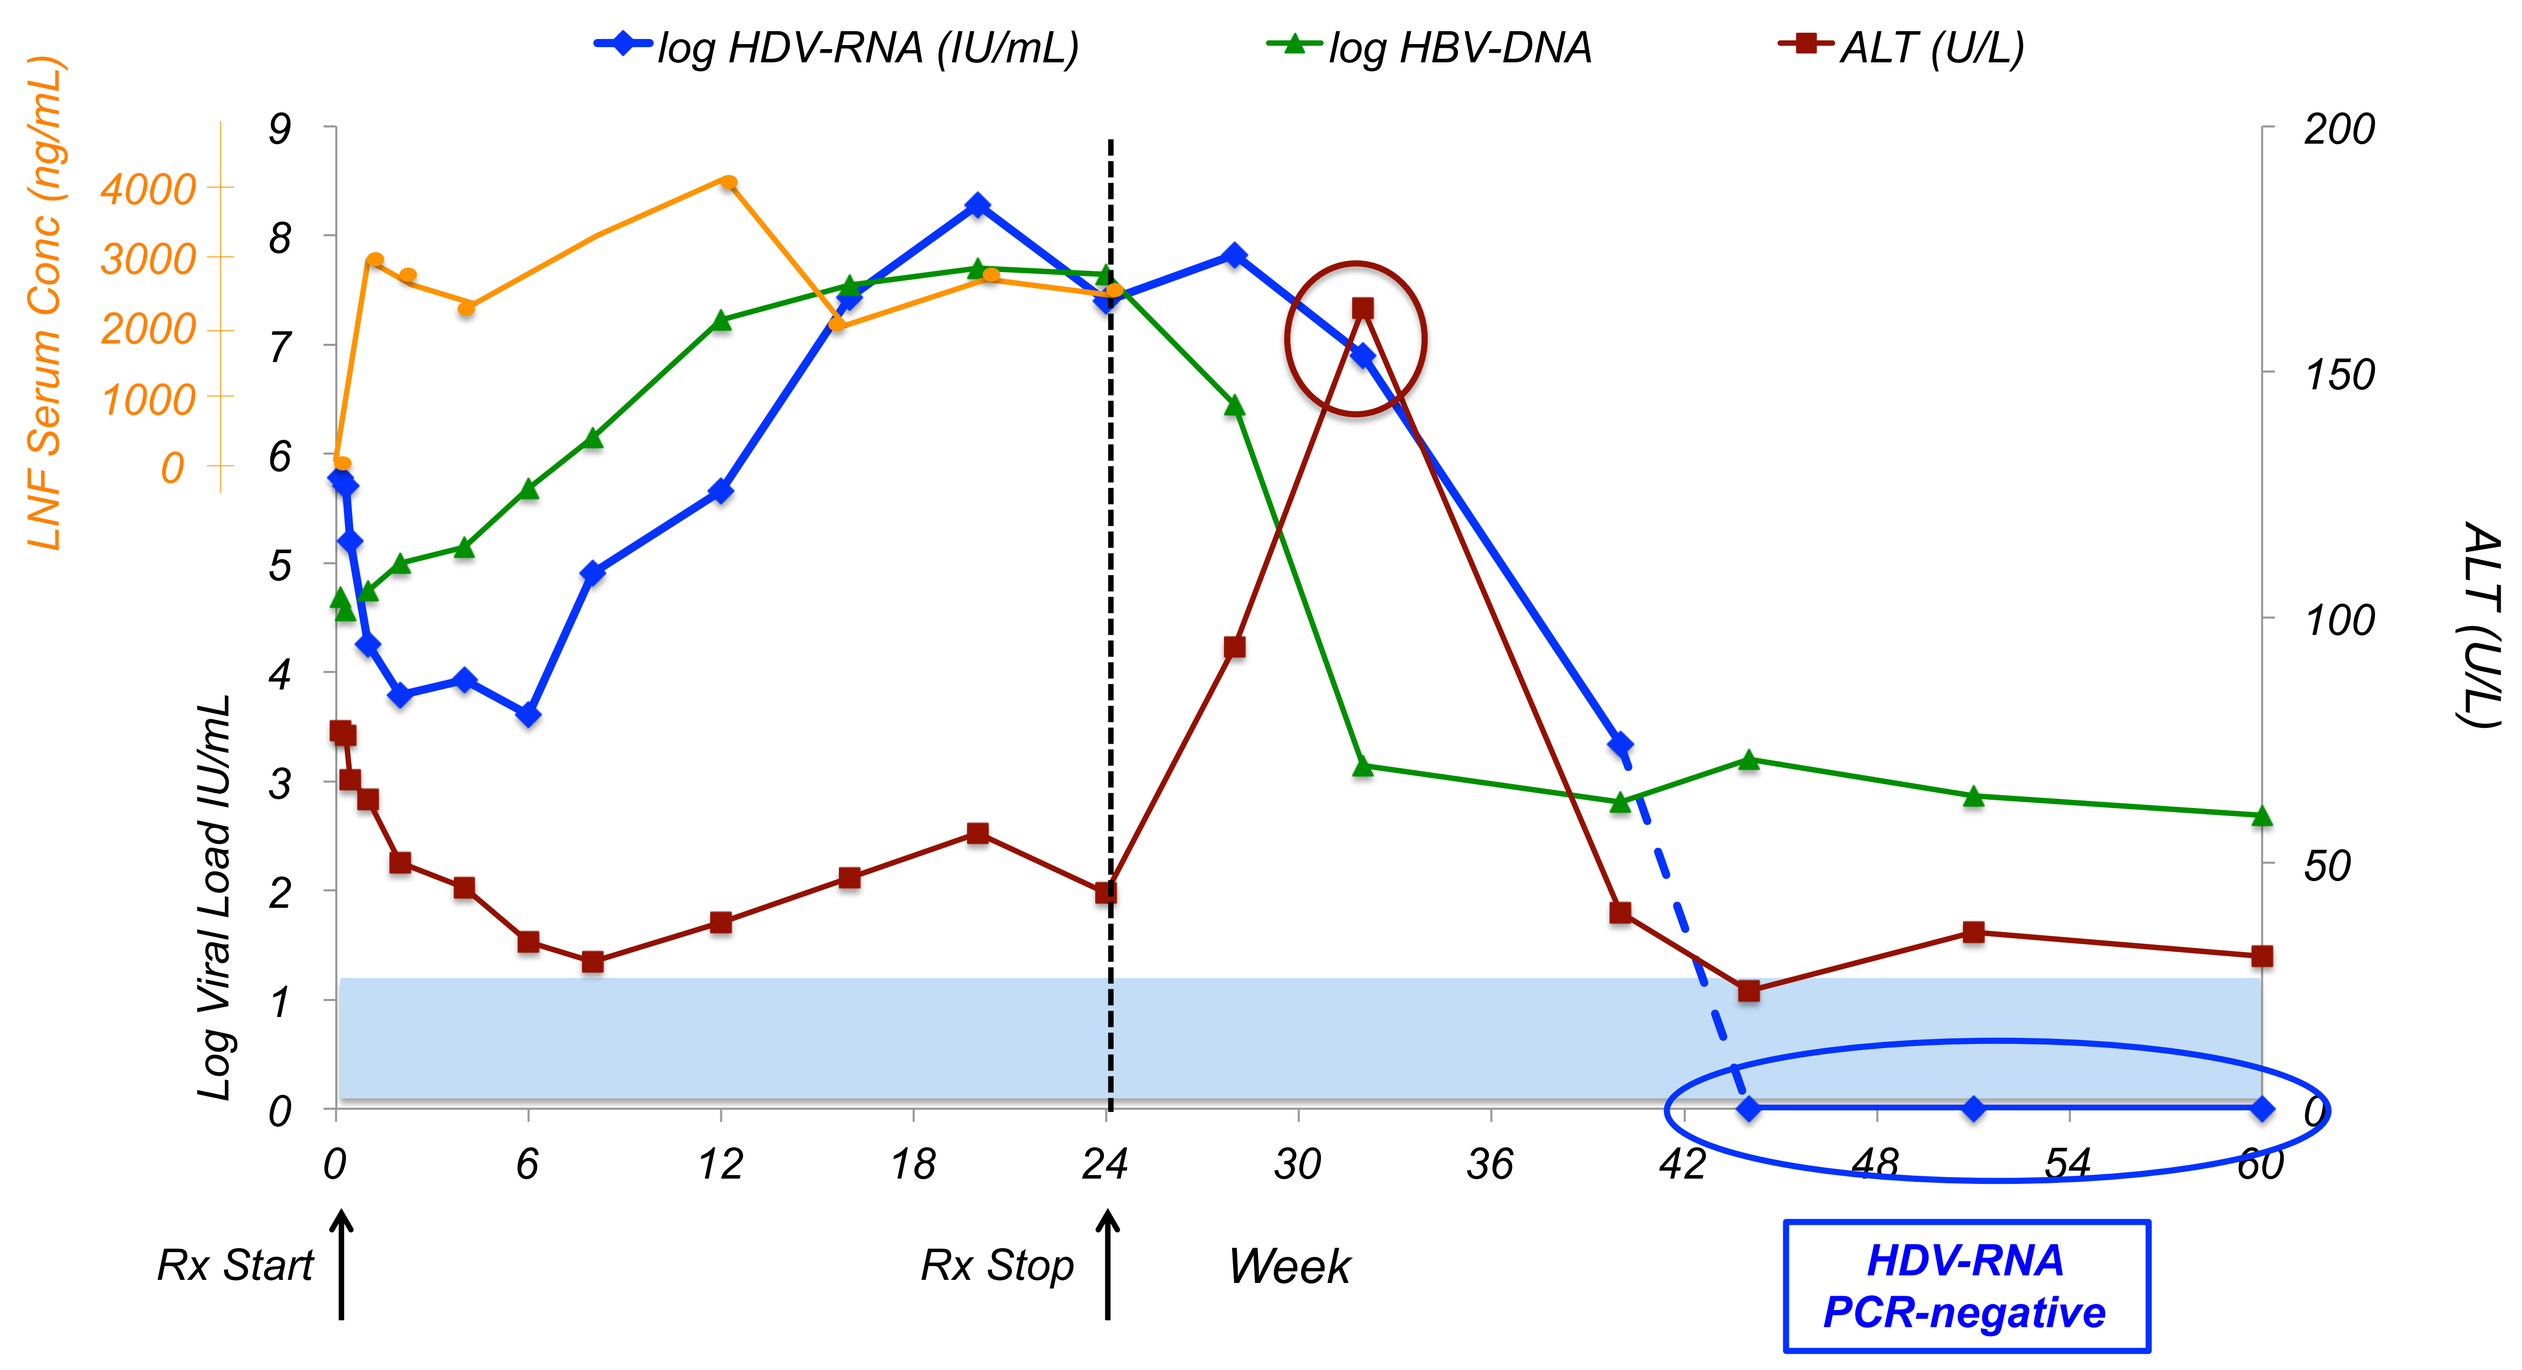

log HDV-RNA (IU/mL)
log HBV-DNA
ALT (U/L)
LNF Serum Conc (ng/mL)
Log Viral Load IU/mL
ALT (U/L)
Week
Rx Start
Rx Stop
HDV-RNA PCR-negative

Patient 3
2 Point Improvement in Fibrosis

A.

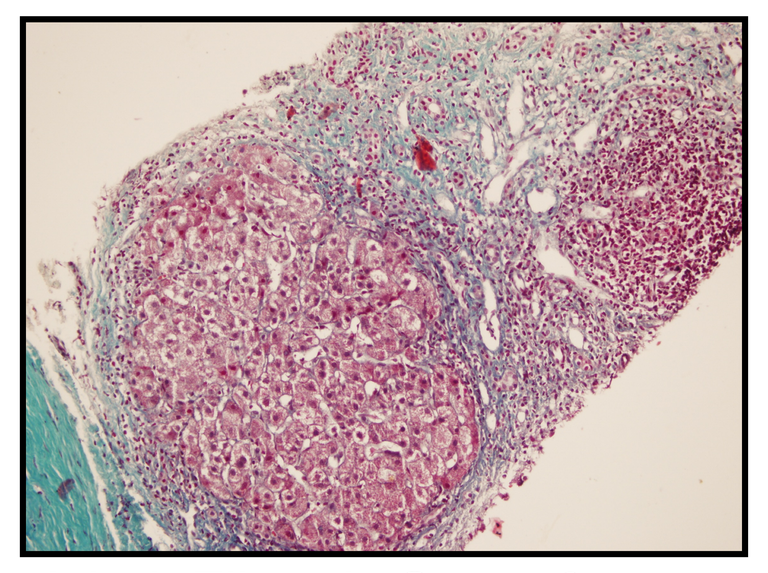

Ishak Fibrosis Score 6

B.

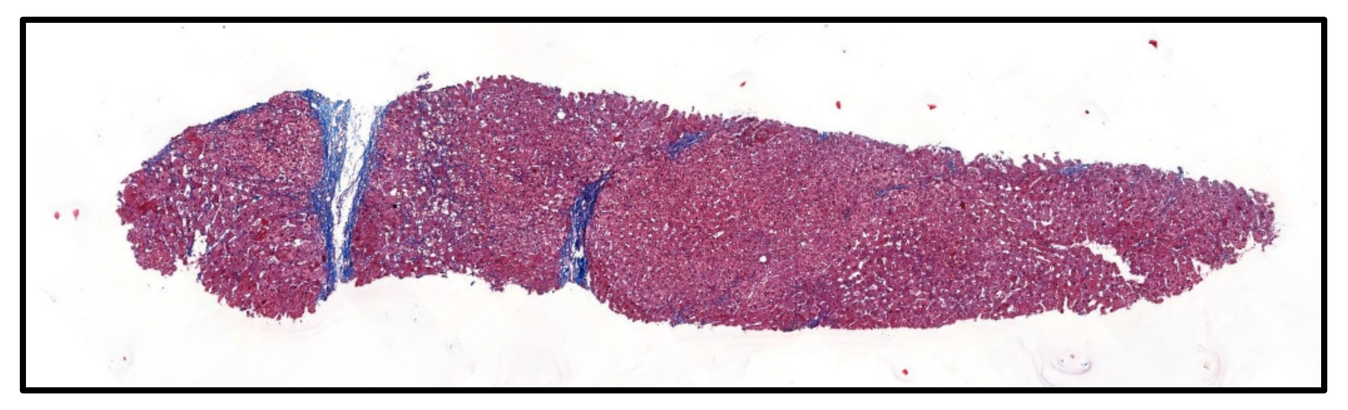

Ishak Fibrosis Score 4

A. Baseline

B. Week 84: 6 months post-ALT normalization and HDV-RNA negativity

4. Results (Continued)

Patient 4
LNF 75 mg BID + RTV 100 mg BID

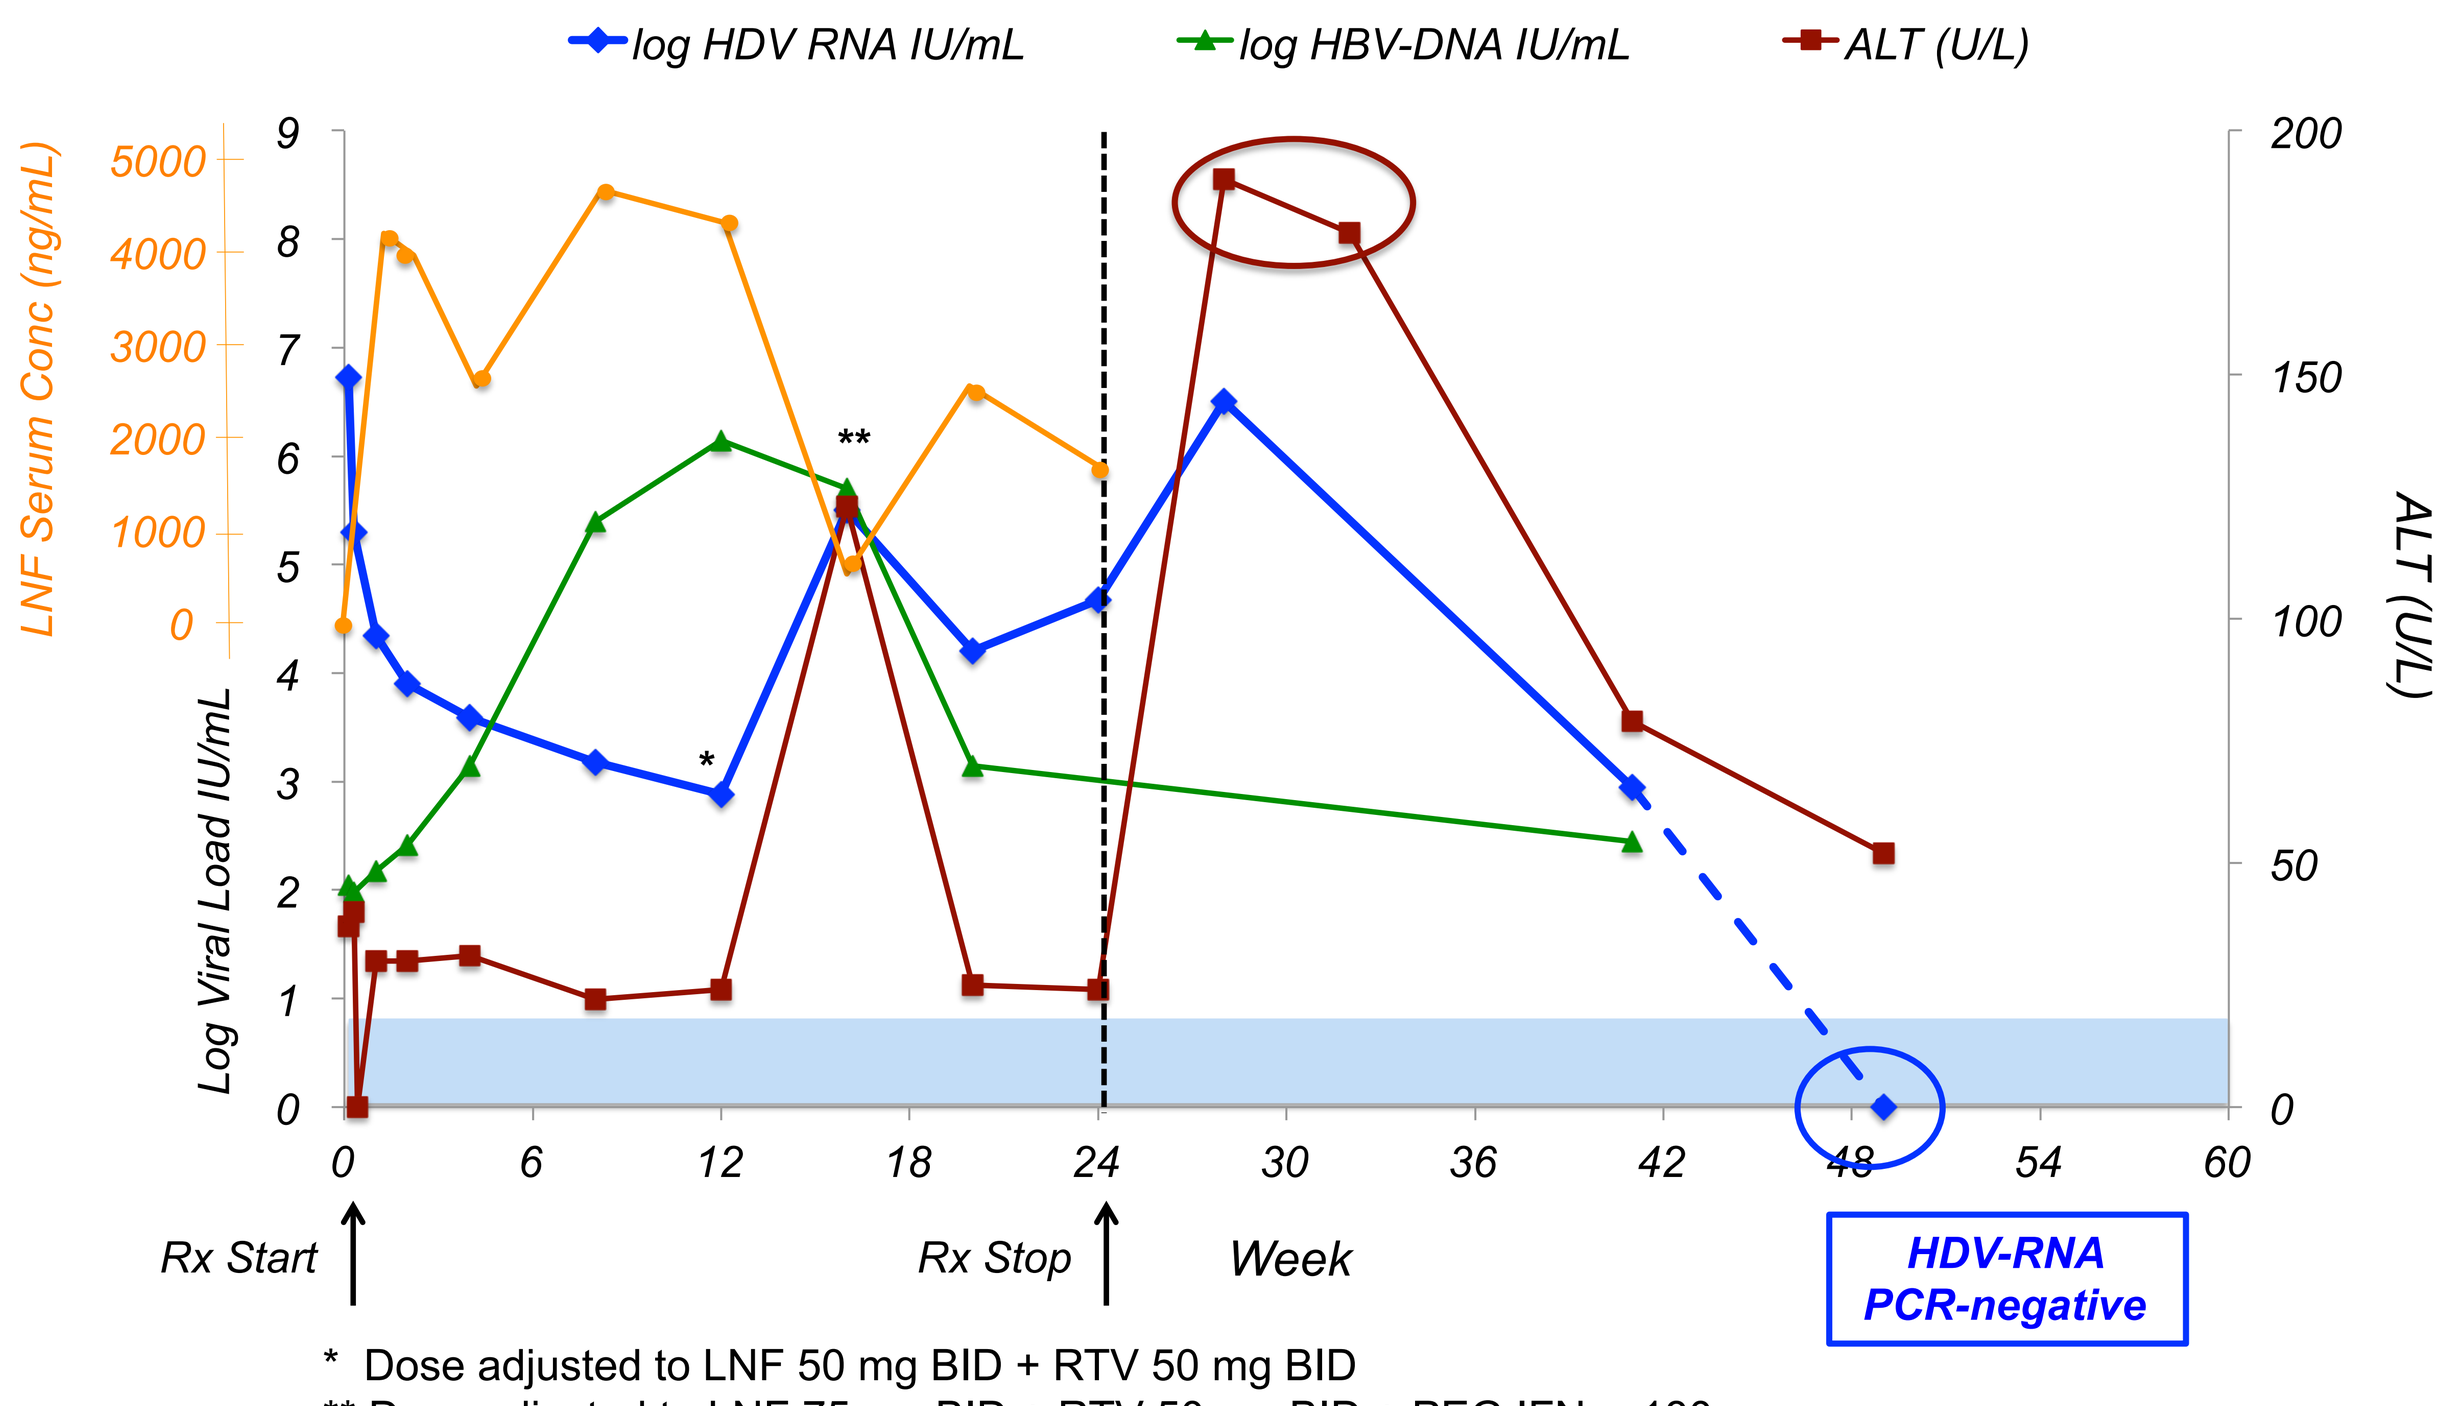

log HDV RNA IU/mL
log HBV-DNA IU/mL
ALT (U/L)
LNF Serum Conc (ng/mL)
Log Viral Load IU/mL
ALT (U/L)
Week
Rx Start
Rx Stop
HDV-RNA PCR-negative

* Dose adjusted to LNF 50 mg BID + RTV 50 mg BID
** Dose adjusted to LNF 75 mg BID + RTV 50 mg BID + PEG IFN-α 180 mcg

Patient 5
LNF 300 mg BID

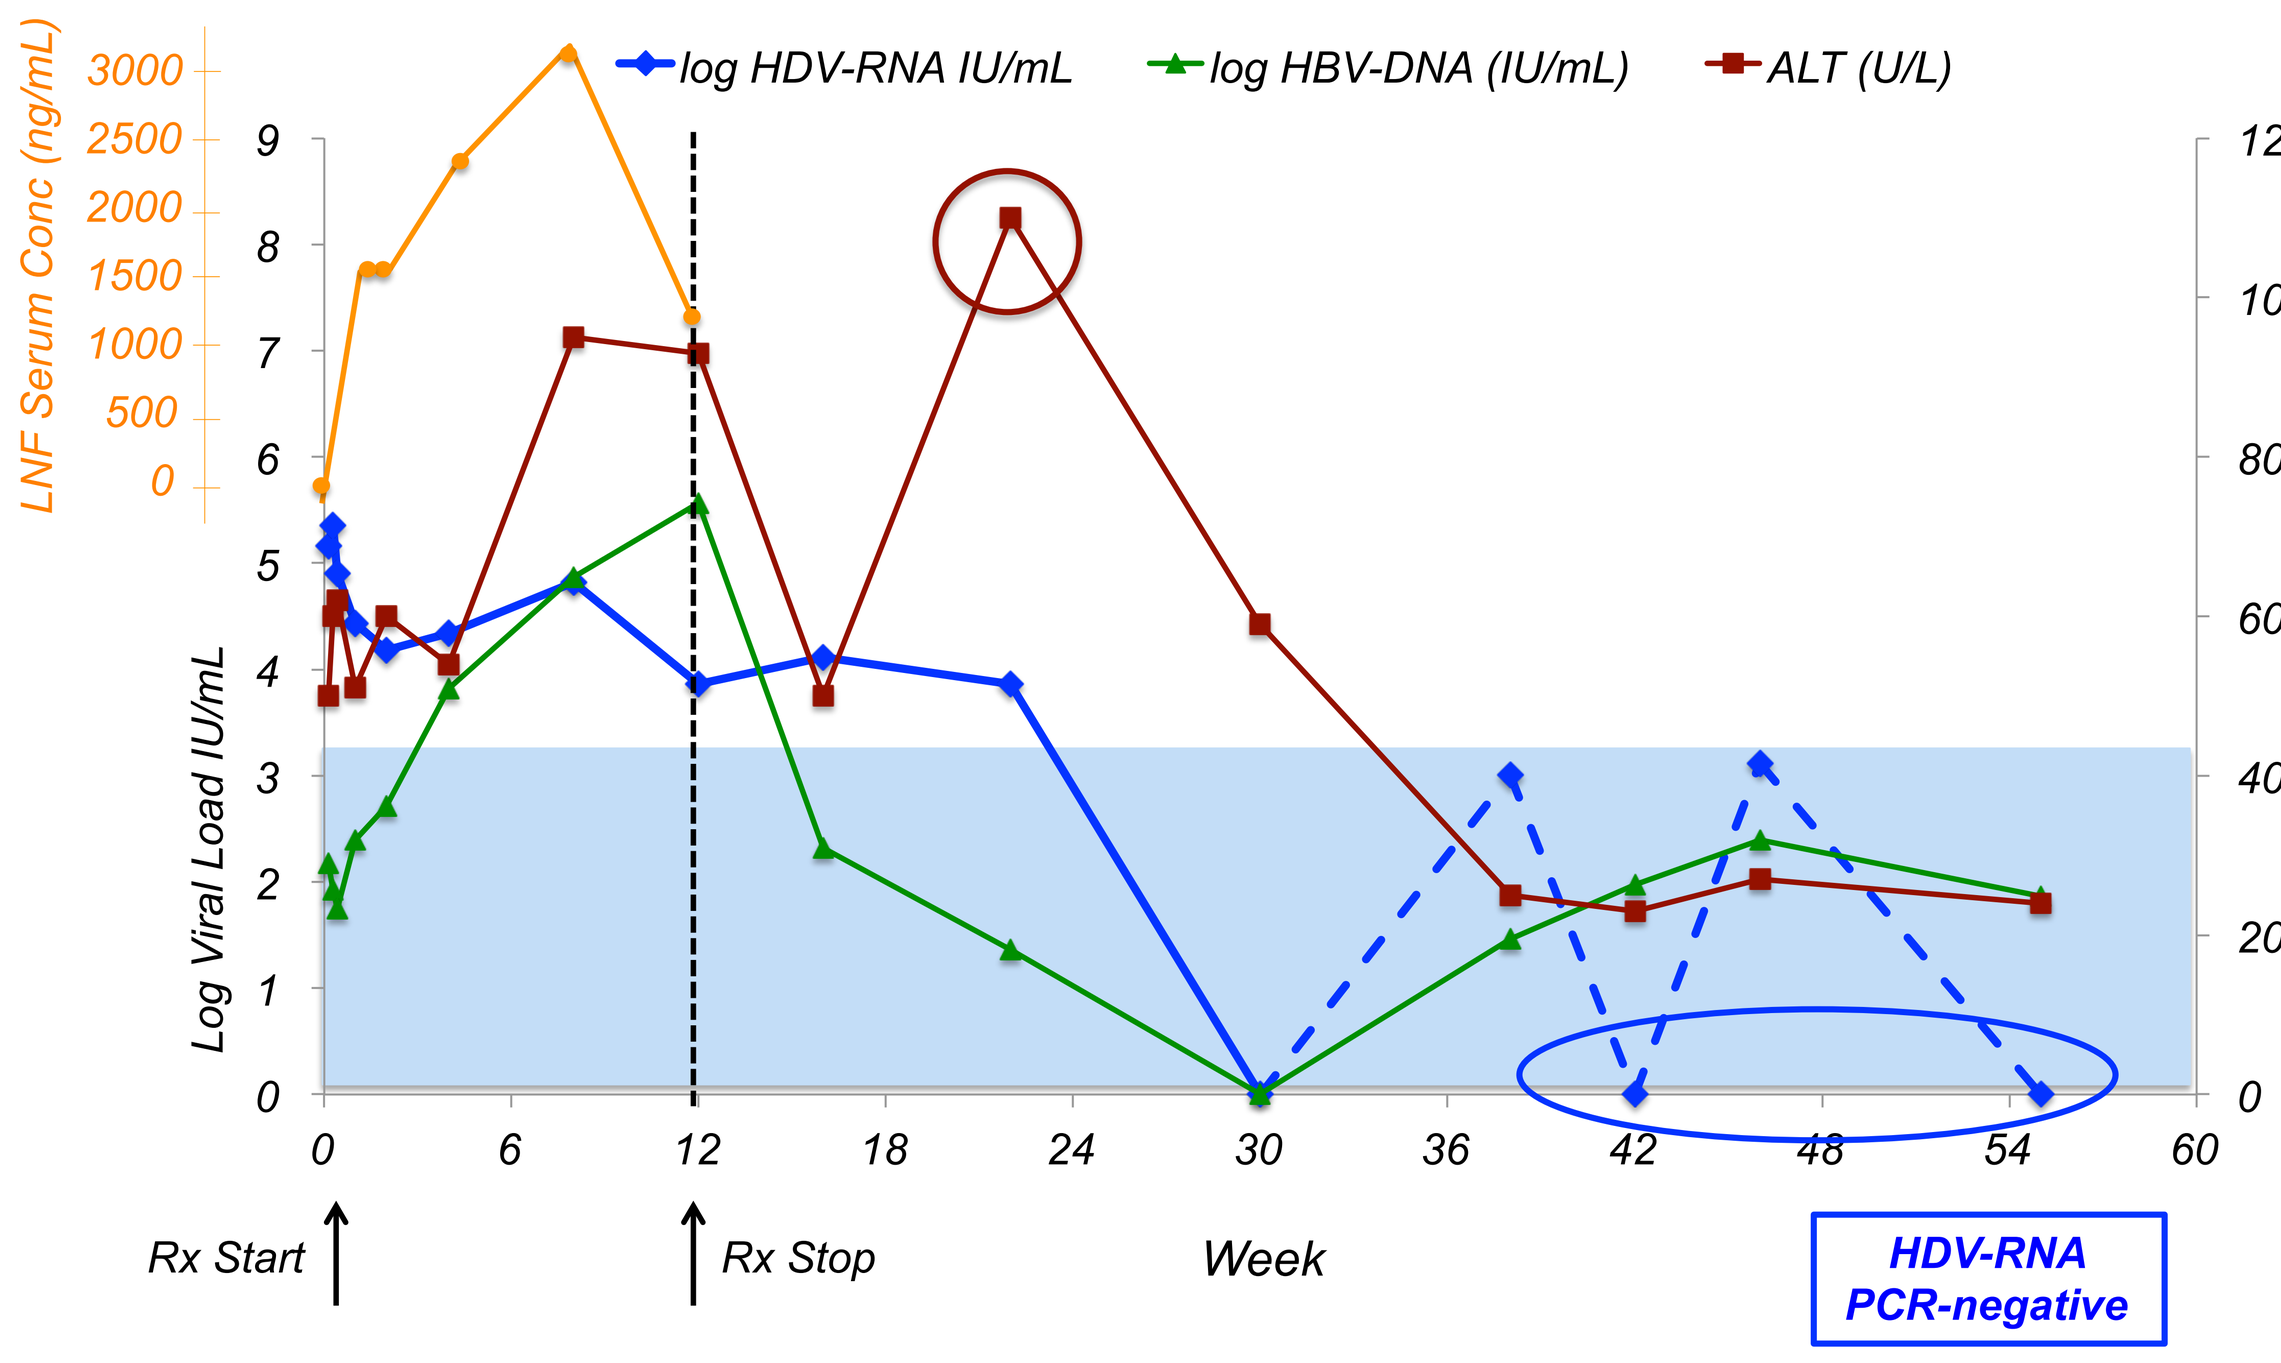

log HDV-RNA IU/mL
log HBV-DNA (IU/mL)
ALT (U/L)
LNF Serum Conc (ng/mL)
Log Viral Load IU/mL
ALT (U/L)
Week
Rx Start
Rx Stop
HDV-RNA PCR-negative

Patient 5
2 Point Improvement in Fibrosis

A.

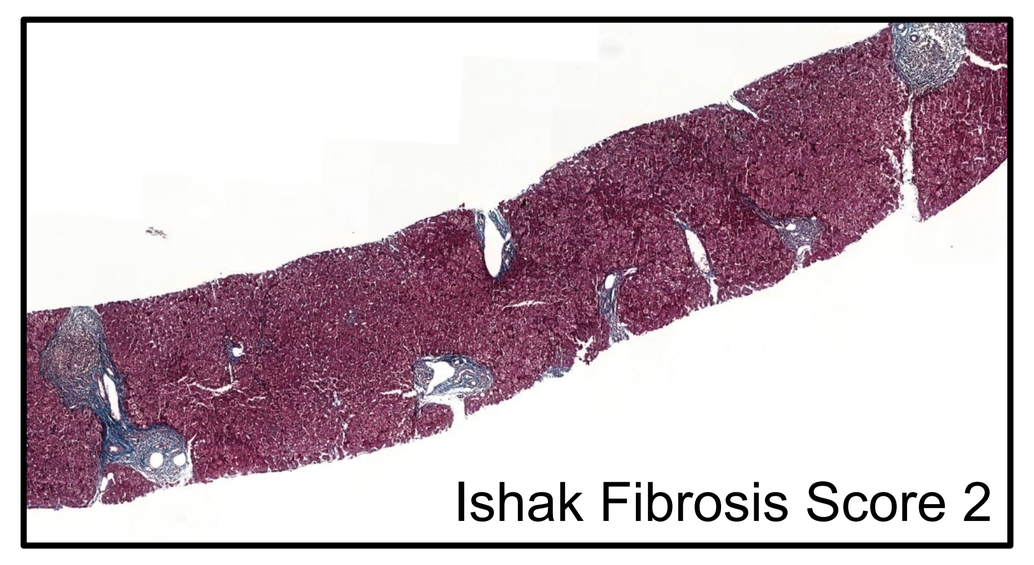

Ishak Fibrosis Score 2

B.

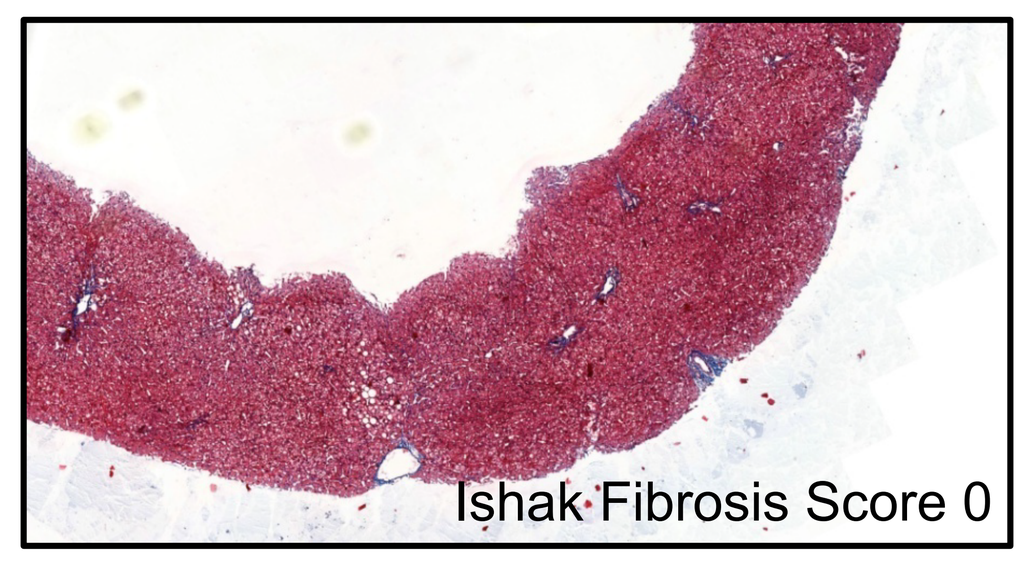

Ishak Fibrosis Score 0

A. Baseline

B. Week 128: 18 months post-ALT normalization and HDV-RNA negativity

5. Conclusions

- Post-treatment ALT flares observed in a subset of HDV-treated patients
 - 5 of 27 (18.5%) patients experienced post-treatment flares
- Post-treatment ALT flares are followed by:
 - HDV-RNA PCR-negativity / HBV-DNA suppression
 - Normalizing ALT levels
- Novel observation (vs. prior IFN studies) in HDV
- LNF treatment may reset and activate immune reactivity to HDV
- Immune reactivity may be spread to HBV (HBV-DNA PCR-negativity)
- One patient experienced transient hyperbilirubinemia associated with the ALT flare, followed by HBsAg to less than 10 IU/mL, ALT normalization, and HDV-RNA PCR negativity (Patient 2)
- Long term ALT normalization has resulted in reversal of fibrosis
- Two potential pathways for achieving HDV-RNA PCR-negativity with LNF therapy:
 - On-treatment LNF-induced HDV-RNA suppression
 → More classical antiviral approach, e.g. ongoing LOWR HDV – 2 study*
 - Post-treatment LNF-induced anti-HDV ALT flares

6. References

*Yurdaydin, C. et al; 2016 AASLD Abstract #1845.

7. Disclosures

Lonafarnib was provided by Eiger Biopharmaceuticals, Inc. Glenn: Equity interest in Eiger Biopharmaceuticals, Inc. Yurdaydin: Received partial travel support from Eiger BioPharmaceuticals, Inc. to attend scientific meetings. All other authors have no financial disclosures.